An induced Pluripotent Stem Cell (iPSC) Vaccine Decreases Tumor Growth and Improves Survival in a Therapeutic Mouse Model of Colon Cancer

Matthias Hundt, Michelle Li, Hui Huang, Carlos A. Obejero-Paz, Peter F. Bove, Ivan Hernandez, Pratima Kundu, Nigel G. Kooreman, Stephen D. Wolpe, Lynne A. Bui

Khloris Biosciences, Mountain View, CA, United States

NOV. 10–14 • WASHINGTON, D.C.

Abstract 775

ABSTRACT

Extensive data on gene expression, metabolic state and glycosylation of cancer cells suggest that cancer represents a reversion of adult cells to an embryonic state and that induced pluripotent stem cells (iPSC) are good surrogates for this state. In contrast to cancer cells, iPSC have never undergone immunoediting and therefore present hundreds of oncofetal antigens in their native conformations. In this study, we administered syngeneic iPSC together with the Toll-like receptor (TLR) 9 agonist CpG1826 in a therapeutic mouse model of colon cancer with and without checkpoint inhibition.

C57BL/6 mice (n=10) were injected with 5×10^5 MC-38 murine colon adenocarcinoma cells s.c. One week later, mice received a course of 4 weekly injections with 10×10^6 irradiated (60 Gy) iPSC admixed with 1 nmol CpG1826, or PBS or CpG alone as controls. Some groups also received anti-PD-1 (clone RMP1-14, 200 µg, 2x/week, i.p.) for 4 weeks. Tumor growth and survival were monitored for 108 days post tumor injection. Mice were euthanized when tumor volume reached 2000 mm^3. Serum IgG binding to iPSC and MC-38 was measured by flow cytometry 1 week after the last immunization in all 60 mice and IFN-γ ELISPOTs were determined after *in vitro* challenge of splenocytes with iPSC and MC-38 lysates in 10 surviving mice at the end of the study.

Treatment of mice with anti-PD-1, anti-PD-1+CpG, iPSC+CpG and iPSC+CpG+anti-PD-1 significantly increased median survival in comparison to CpG alone (Gehan-Breslow-Wilcoxon test) by ~50% (Table below). Therapeutic vaccination with iPSC+CpG was as effective as treatment with anti-PD-1, and anti-PD-1+CpG. Similar data were obtained for tumor growth; iPSC+CpG vaccination was as effective in reducing tumor growth as anti-PD-1. Only immunization with iPSC+CpG and iPSC+CpG+anti-PD-1 induced a significant increase in serum IgG binding to iPSC and MC-38; IFN-γ spots after *in vitro* challenge with iPSC and MC-38 lysates were only detectable in mice that had been injected with iPSC.

Irradiated syngeneic iPSC admixed with TLR9 agonist CpG1826 with or without combination with checkpoint inhibition induced T cell and antibody responses to iPSC that cross-reacted with MC-38. The iPSC vaccine was effective in delaying and decreasing tumor growth and in increasing median survival in a therapeutic model of colon cancer comparable to checkpoint inhibition.

Treatment	PBS	CpG	αPD-1	CpG+αPD-1	iPSC+CpG	iPSC+CpG+αPD-1
Median survival [days]	58	52	82.5	74.5	73.5	75.5
P value for CpG vs ...	0.9697	NA	0.0332	0.0107	0.0269	0.0156

STUDY DESIGN

MC-38 sc Colon Cancer Mouse Model

Group	# of mice	Treatment Groups	Dose	Tumor Cell
1	n=10	PBS (sc)	control	MC-38 Adenocarcinoma 500,000 cells; sc injection
2	n=10	CpG (sc)	1 nmol	
3	n=10	anti-PD-1 (ip)	200 µg 2x/wk.	
4	n=10	CpG + anti-PD-1	1 nmol/ 200 µg 2x/wk.	
5	n=10	iPSC + CpG (sc)	10×10^6 cells / 1 nmol	
6	n=10	iPSC + CpG + anti-PD-1	10×10^6 cells / 1 nmol / 200 µg 2x/wk	

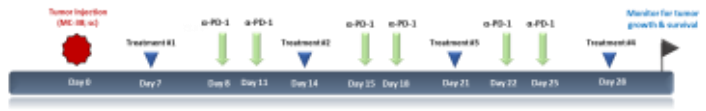
Tumor injection (MC-38, sc)
Treatment #1
Treatment #2
Treatment #3
Treatment #4
α-PD-1
Monitor for tumor growth & survival
Day 0
Day 7
Day 8
Day 11
Day 14
Day 15
Day 18
Day 21
Day 22
Day 25
Day 28

TUMOR GROWTH

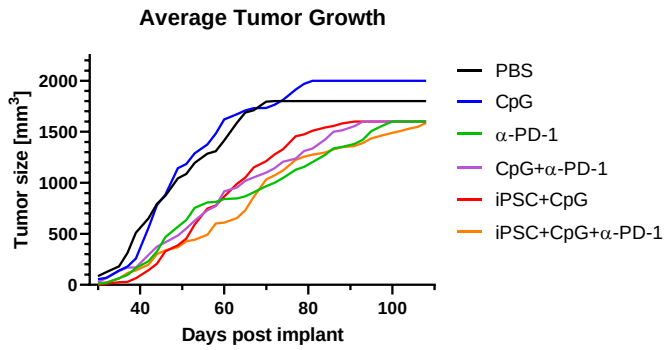
Average Tumor Growth
Tumor size [mm³]
Days post implant
PBS
CpG
α-PD-1
CpG+α-PD-1
iPSC+CpG
iPSC+CpG+α-PD-1

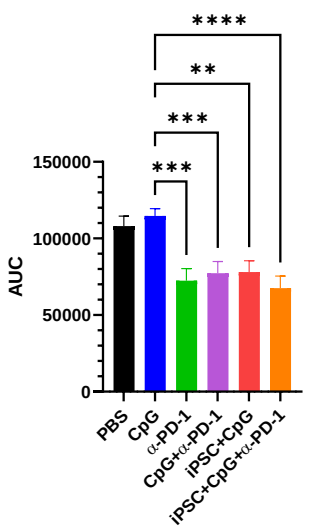
AUC

**

PBS
CpG
α-PD-1
CpG+α-PD-1
iPSC+CpG
iPSC+CpG+α-PD-1

After subcutaneous injection of MC-38 cells into the right flank tumor size was measured at least twice a week using a caliper. Tumor volume was calculated as $V = LxW^2$.

Therapeutic treatment with anti-PD-1, CpG+anti-PD-1, iPSC+CpG and iPSC+CpG+anti-PD-1 significantly delayed tumor growth in comparison to treatment with CpG alone as assessed by calculation of the area under the curve (AUC) of the increase of tumor volume over time (108 days). The effect of iPSC+CpG was as strong as checkpoint inhibition. One-way ANOVA with Dunnett's multiple comparison test (**, $p<0.01$; ***, $p<0.001$; ****, $p<0.0001$).

SURVIVAL

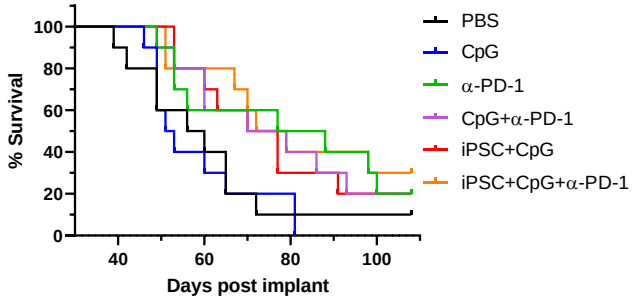
% Survival
Days post implant
PBS
CpG
α-PD-1
CpG+α-PD-1
iPSC+CpG
iPSC+CpG+α-PD-1

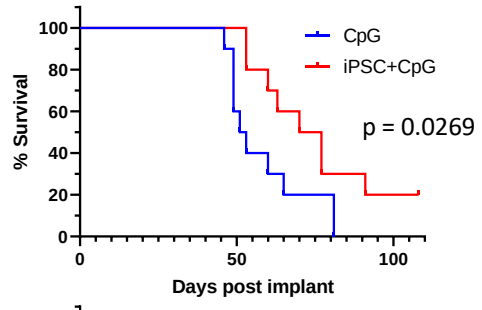
% Survival
Days post implant
CpG
iPSC+CpG
p = 0.0269

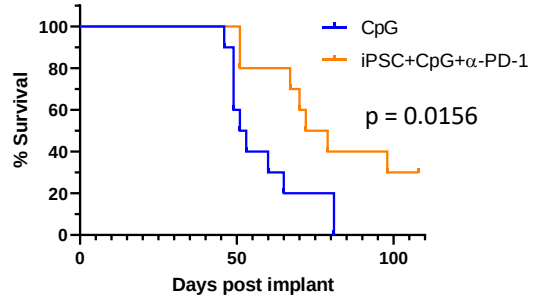
% Survival
Days post implant
CpG
iPSC+CpG+α-PD-1
p = 0.0156

Treatment of mice with iPSC+CpG and iPSC+CpG+anti-PD-1 significantly increased survival of mice in comparison to treatment with CpG alone as determined by Gehan-Breslow-Wilcoxon test.

REFERENCES

Kooreman NG, Kim Y, de Almeida PE, et al. Autologous iPSC-Based Vaccines Elicit Anti-tumor Responses In Vivo. Cell Stem Cell. 2018;22(4):501-513.e7. doi:10.1016/j.stem.2018.01.016

Wang L, Pegram MD, Wu JC. Induced pluripotent stem cells as a novel cancer vaccine. Expert Opin Biol Ther. 2019;19(11):1191-1197. doi:10.1080/14712598.2019.1650909

Ouyang X, Liu Y, Zhou Y, et al. Antitumor effects of iPSC-based cancer vaccine in pancreatic cancer. Stem Cell Reports. 2021;16(6):1468-1477. doi:10.1016/j.stemcr.2021.04.004

Ghosh Z, Huang M, Hu S, Wilson KD, Dey D, Wu JC. Dissecting the oncogenic and tumorigenic potential of differentiated human induced pluripotent stem cells and human embryonic stem cells. Cancer Res. 2011;71(14):5030-5039. doi:10.1158/0008-5472.CAN-10-4402

HUMORAL IMMUNE RESPONSE

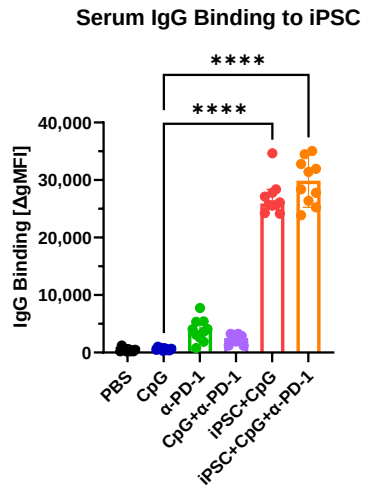
Serum IgG Binding to iPSC
IgG Binding [ΔgMFI]

PBS
CpG
α-PD-1
CpG+α-PD-1
iPSC+CpG
iPSC+CpG+α-PD-1

Serum from all 60 mice was obtained one week after the last (4th) treatment.

Serum (1:50 dilution) was incubated with iPSC and MC-38 and binding measured with IgG-specific antiserum by flow cytometry.

Only treatments containing iPSC induced significant serum IgG binding to iPSC. Kruskal-Wallis test with Dunnett's multiple comparison test (****, $p<0.0001$).

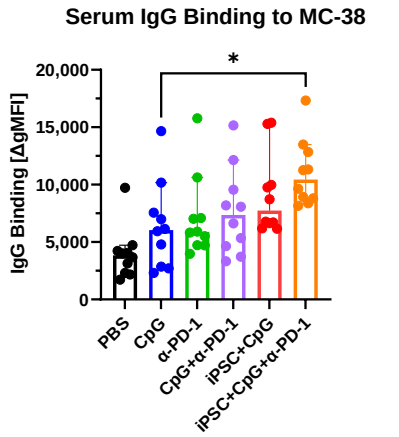
Serum IgG Binding to MC-38
IgG Binding [ΔgMFI]
*
PBS
CpG
α-PD-1
CpG+α-PD-1
iPSC+CpG
iPSC+CpG+α-PD-1

Binding to MC-38 was significantly increased after treatment iPSC+CpG+anti-PD-1 in comparison to CpG. Kruskal-Wallis test with Dunnett's multiple comparison test (*, $p<0.05$).

CONTACT

Khloris Biosciences
Matthias Hundt
319 N Bernardo Ave
Mountain View, CA 94043
United States
mhundt@khlorisbiosciences.com

CELLULAR IMMUNE RESPONSE

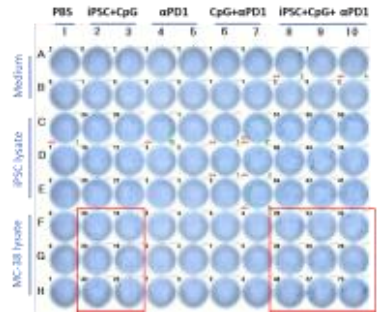
PBS
iPSC+CpG
αPD1
CpG+αPD1
iPSC+CpG+ αPD1
Medium
iPSC lysate
MC-38 lysate

Ten out of 60 mice were alive on day 108. Nine of these 10 mice were tumor-free and one had a measurable tumor (#9, iPSC+CpG+anti-PD-1 group). These mice were euthanized to obtain splenocytes to perform an IFN-γ ELISPOT assay. 500,000 splenocytes per well were challenged *in vitro* with 35 µg lysate for 20 hours.

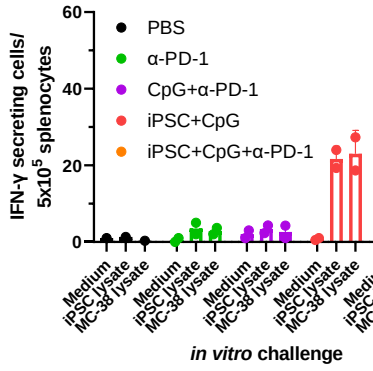
IFN-γ secreting cells/ 5x10⁵ splenocytes
PBS
α-PD-1
CpG+α-PD-1
iPSC+CpG
iPSC+CpG+α-PD-1
Medium
iPSC lysate
MC-38 lysate
in vitro challenge

CONCLUSIONS

A mouse model of colon cancer with sensitivity to checkpoint inhibition was chosen to investigate the efficacy of a syngeneic iPSC vaccine and its immunogenicity.

1. Significant reduction of tumor growth and increase of survival after treatment with iPSC vaccine (iPSC+CpG and iPSC+CpG+anti-PD-1)

2. iPSC+CpG as effective as anti-PD-1 and anti-PD-1+CpG, but no additive or synergistic effect between iPSC vaccine and anti-PD-1

3. Significant induction of iPSC and MC-38 binding antibodies after iPSC vaccination. No induction of iPSC antibodies by anti-PD-1 treatment.

4. T cell response after iPSC vaccination without and with combination with anti-PD-1.

ACKNOWLEDGEMENTS

The study was approved by Valley Bio Services' Institutional Animal Care and Use Committee; approval number VBS1002.