Both Allogeneic and Syngeneic induced Pluripotent Stem Cell (iPSC) Vaccines Decrease Tumor Growth and Improve Survival in a Prophylactic Mouse Model of Breast Cancer

Sahil Inamdar, Carlos A. Obejero-Paz, Pratima Kundu, Skylar R. Foust, Ivan Hernandez, Nigel G. Kooreman, Stephen D. Wolpe, Lynne A. Bui, Matthias Hundt

Khloris Biosciences, San Jose, CA, United States

SITC 2023 San Diego, Nov. 1–5

Abstract 415

Introduction

- Immunotherapies involving induced pluripotent stem cells (iPSCs) are an effective strategy for cancer treatment in preclinical models.
- Extensive data on gene expression, metabolic state and glycosylation of cancer cells suggest that cancer represents a reversion of adult cells to an embryonic state and iPSC model this state.
- In contrast to cancer cells, iPSC have never undergone immunoediting and therefore present hundreds of oncofetal antigens in their native conformations.
- One major advantage of a whole-cell vaccination over other vaccines, which consist of recombinant protein(s) or mRNA, is that a wide range of antigens can be presented to T cells, which also include unknown antigens.
- Previous studies have demonstrated the efficacy of syngeneic iPSC vaccination in a variety of cancer models and robust cellular and humoral immune responses.
- In this study, we administered vaccines comprising syngeneic or allogeneic iPSC together with the Toll-like receptor (TLR) 9 agonist CpG1826 (CpG) as an adjuvant and compared their immunogenicity and preclinical efficacy in a prophylactic mouse model of breast cancer.

Methods

- Female FVB mice, 6-8 weeks old, were injected s.c. every 7 days with 10×10^6 irradiated (60 Gy) iPSC derived from FVB (syngeneic) or C57BL/6 (B6, allogeneic) mice admixed with 1 nmol CpG, or CpG alone as control for a total of 6 vaccinations.
- For tumor inoculation, syngeneic breast cancer DB7 cells were s.c. injected as 0.5×10^6 cells/mouse in 200 µL PBS into the lower right flank.

(A)

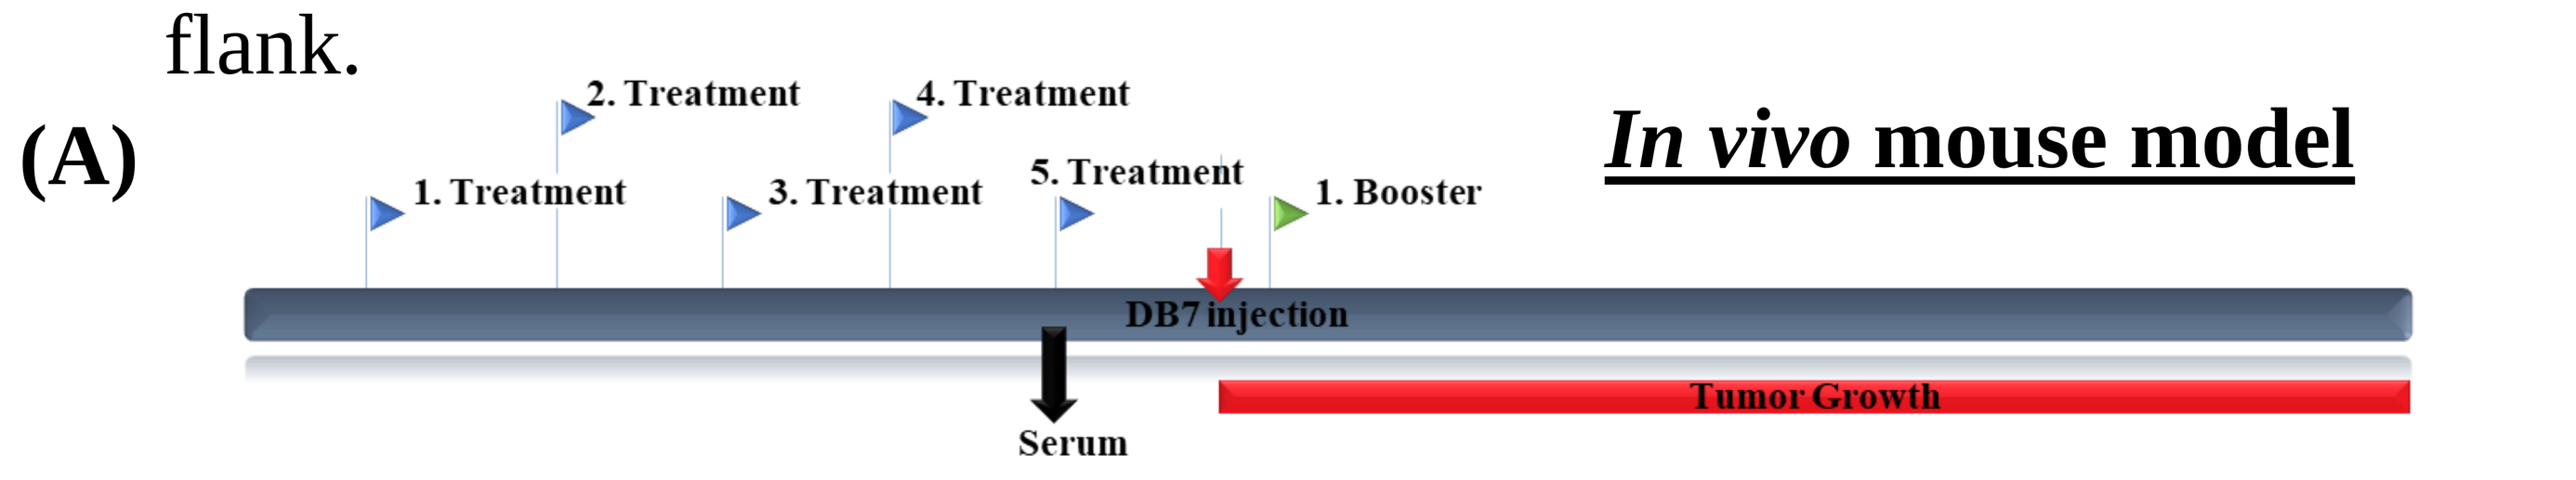
In vivo mouse model
1. Treatment
2. Treatment
3. Treatment
4. Treatment
5. Treatment
1. Booster
DB7 injection
Serum
Tumor Growth

(B)

Mice	Treatment	Dose	Day 0	Day 7	Day 14	Day 21	Day 28	Day 28	Day 35	Day 37	Endpoints
FVB mice (10)	CpG in Cryostor	1 nmol/ mouse	CpG	CpG	CpG	CpG	CpG	Serum 100uL/ mouse	0.5×10^6 DB7 cells SC/mouse	CpG	Tumor size and survival
FVB mice (10)	FVB-iPSC in Cryostor+CpG	10×10^6 + 1 nmol/ mouse	Vac	Vac	Vac	Vac	Vac			Vac	
FVB mice (10)	B6-iPSC in Cryostor+CpG	10×10^6 + 1 nmol/ mouse	Vac	Vac	Vac	Vac	Vac			Vac	

Figure 1: Overall schematic and timeline of the murine breast cancer study, *in vivo*. (A) Timeline of vaccinations for the mouse model. **(B)** Treatment groups studied in this mouse model.

- All mice were randomized and divided into 10 mice/group before inoculation with tumor cells.
- Tumor growth was measured twice weekly using digital caliper and calculated as $V = L \cdot W^2$. Mice were euthanized then the tumor size excided ≥2000 mm³.

Results (*in vivo*)

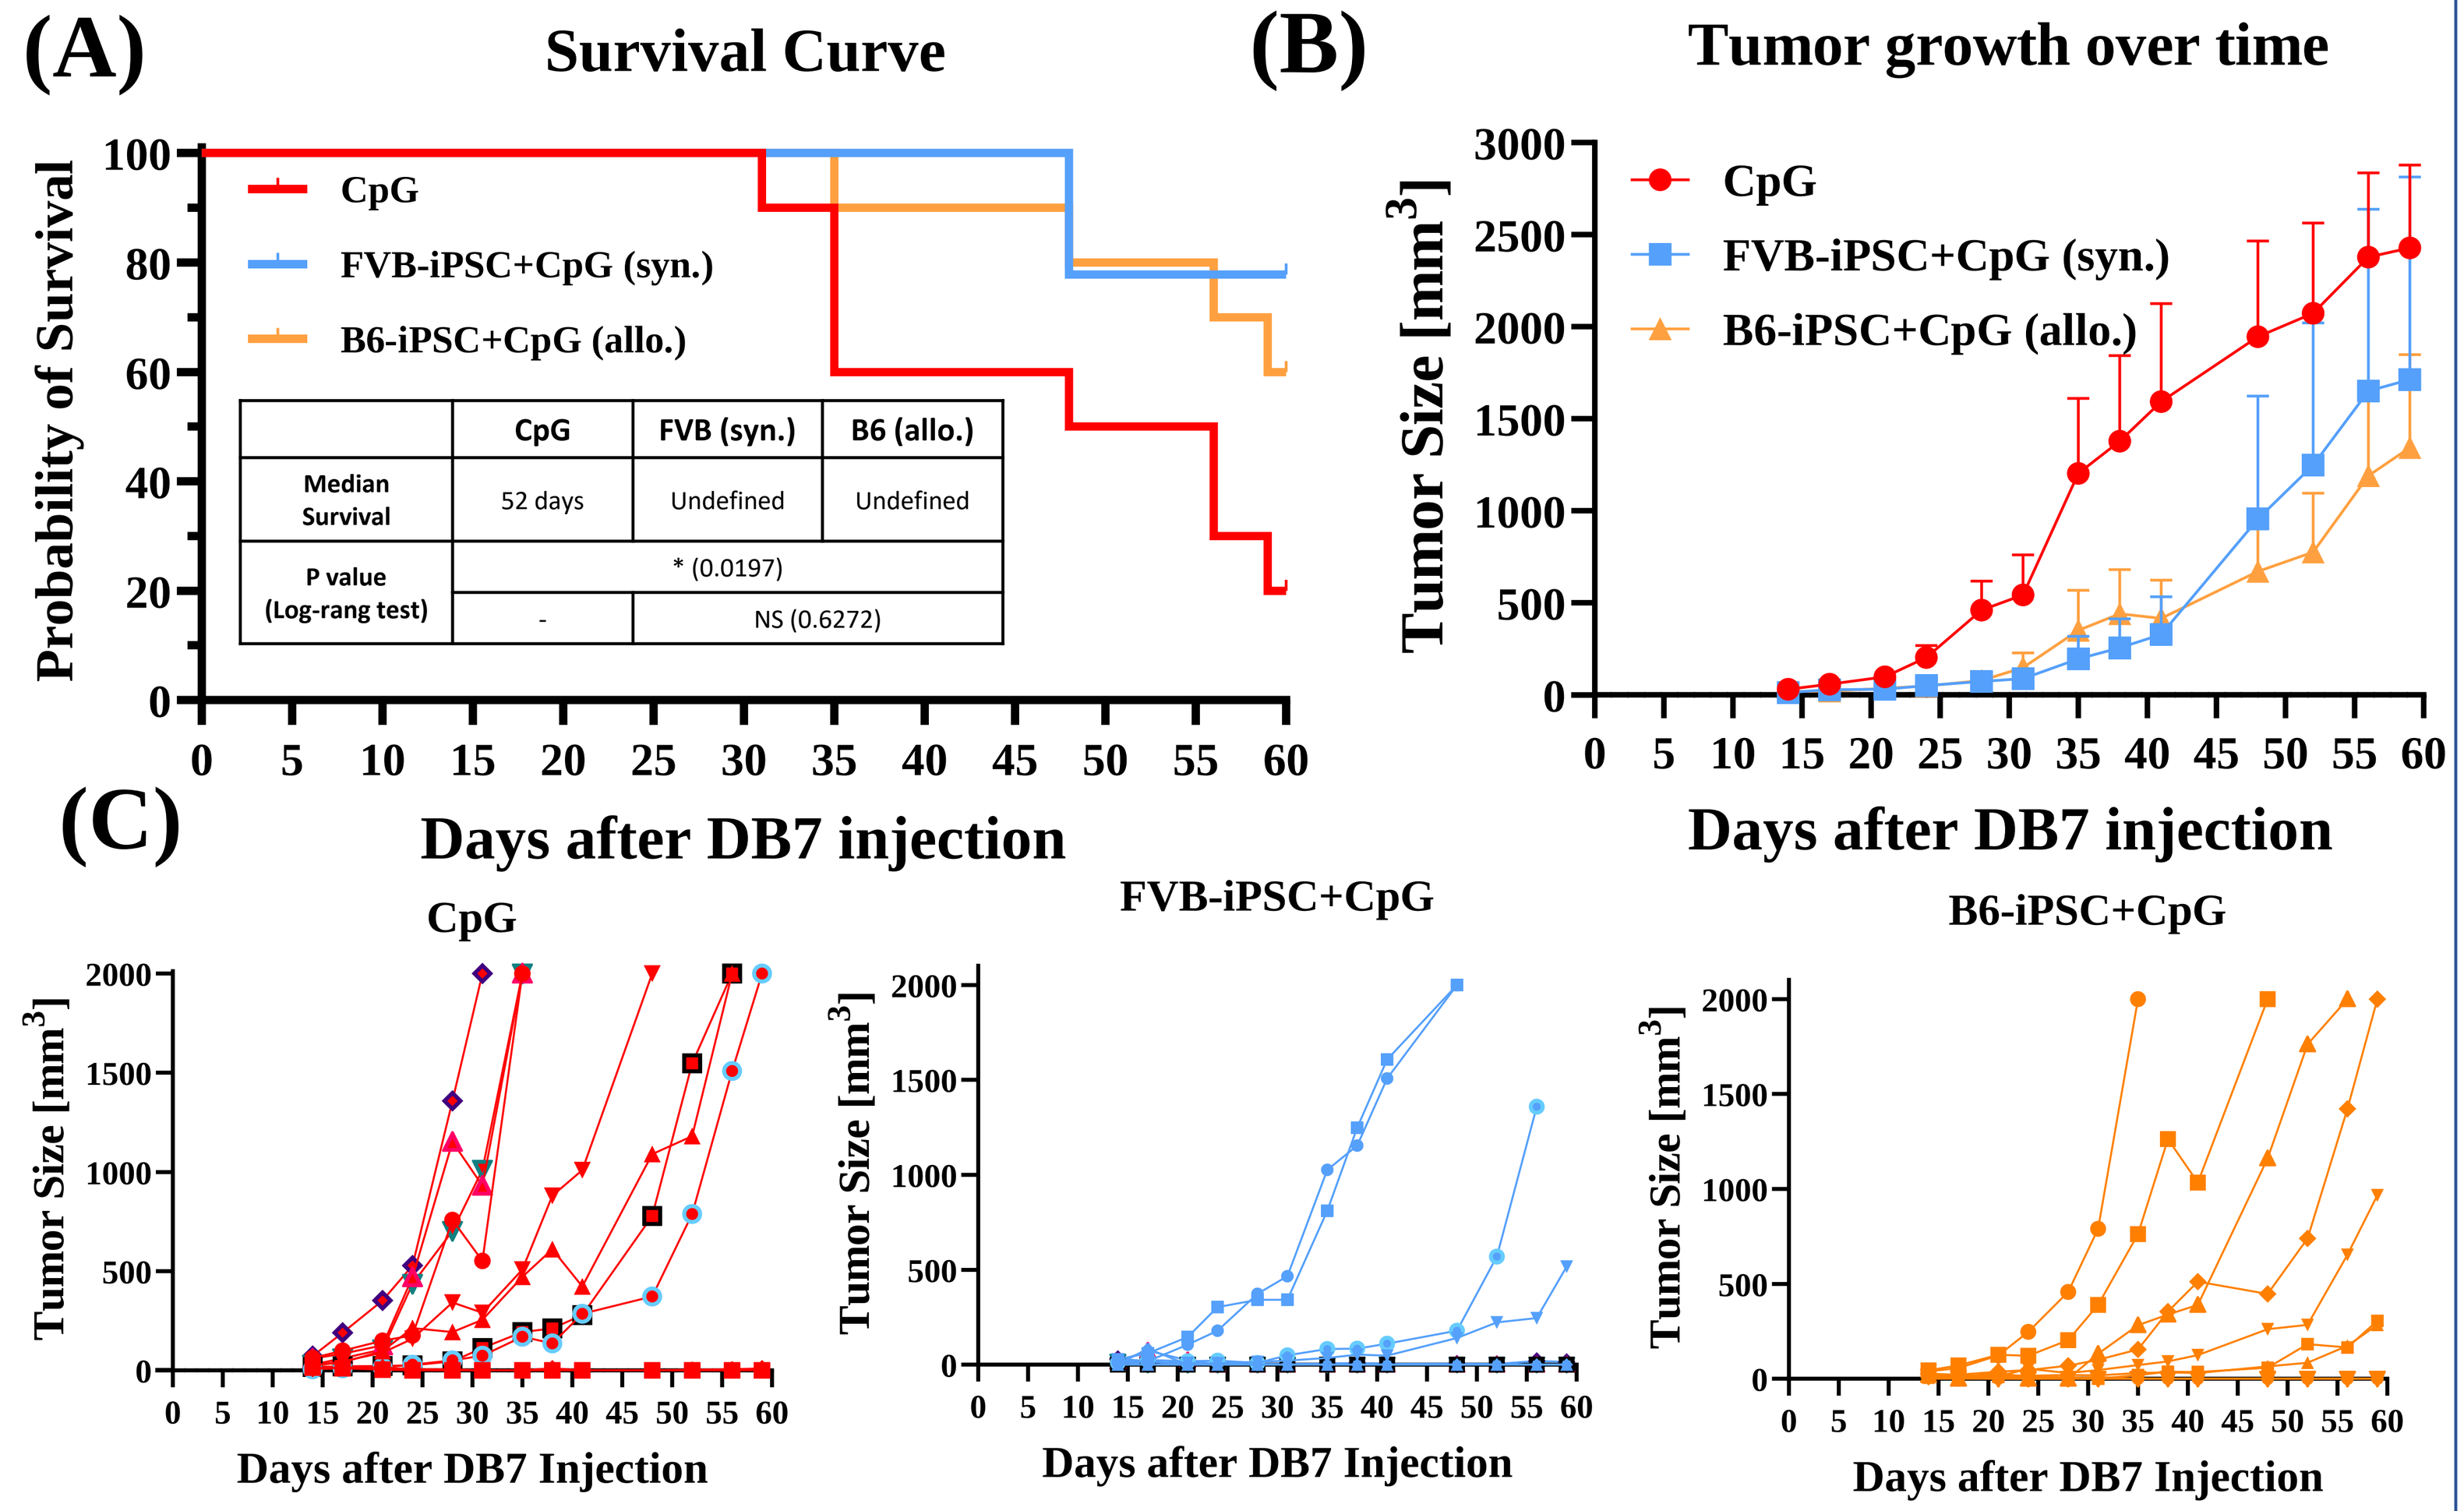
(A) Survival Curve
CpG
FVB-iPSC+CpG (syn.)
B6-iPSC+CpG (allo.)
Probability of Survival
Days after DB7 injection
Median Survival: CpG 52 days; FVB (syn.) Undefined; B6 (allo.) Undefined
P value (Log-rang test): * (0.0197); - ; NS (0.6272)
(B) Tumor growth over time
Tumor Size [mm³]
Days after DB7 injection
(C)
CpG
FVB-iPSC+CpG
B6-iPSC+CpG
Days after DB7 Injection

Figure 2: Allogeneic B6-iPSC vaccine is as efficacious as syngeneic FVB-iPSC vaccine in delaying tumor growth in breast cancer model. (A) Kaplan- Meier curve indicating similar survival of syngeneic-iPSC vaccine treated as well as allogeneic-iPSC vaccine treated mice. (Log-rang Mantel-Cox test) **(B)** Average tumor growth in mice throughout the study confirms that there is no difference in tumor growth (p=0.97) between syngeneic and allogeneic iPSC vaccines. (n=10, mean with SEM, One-way ANOVA with Tukey's test of AUC). (C) Tumor growth in individual mice throughout the study

Humoral Response

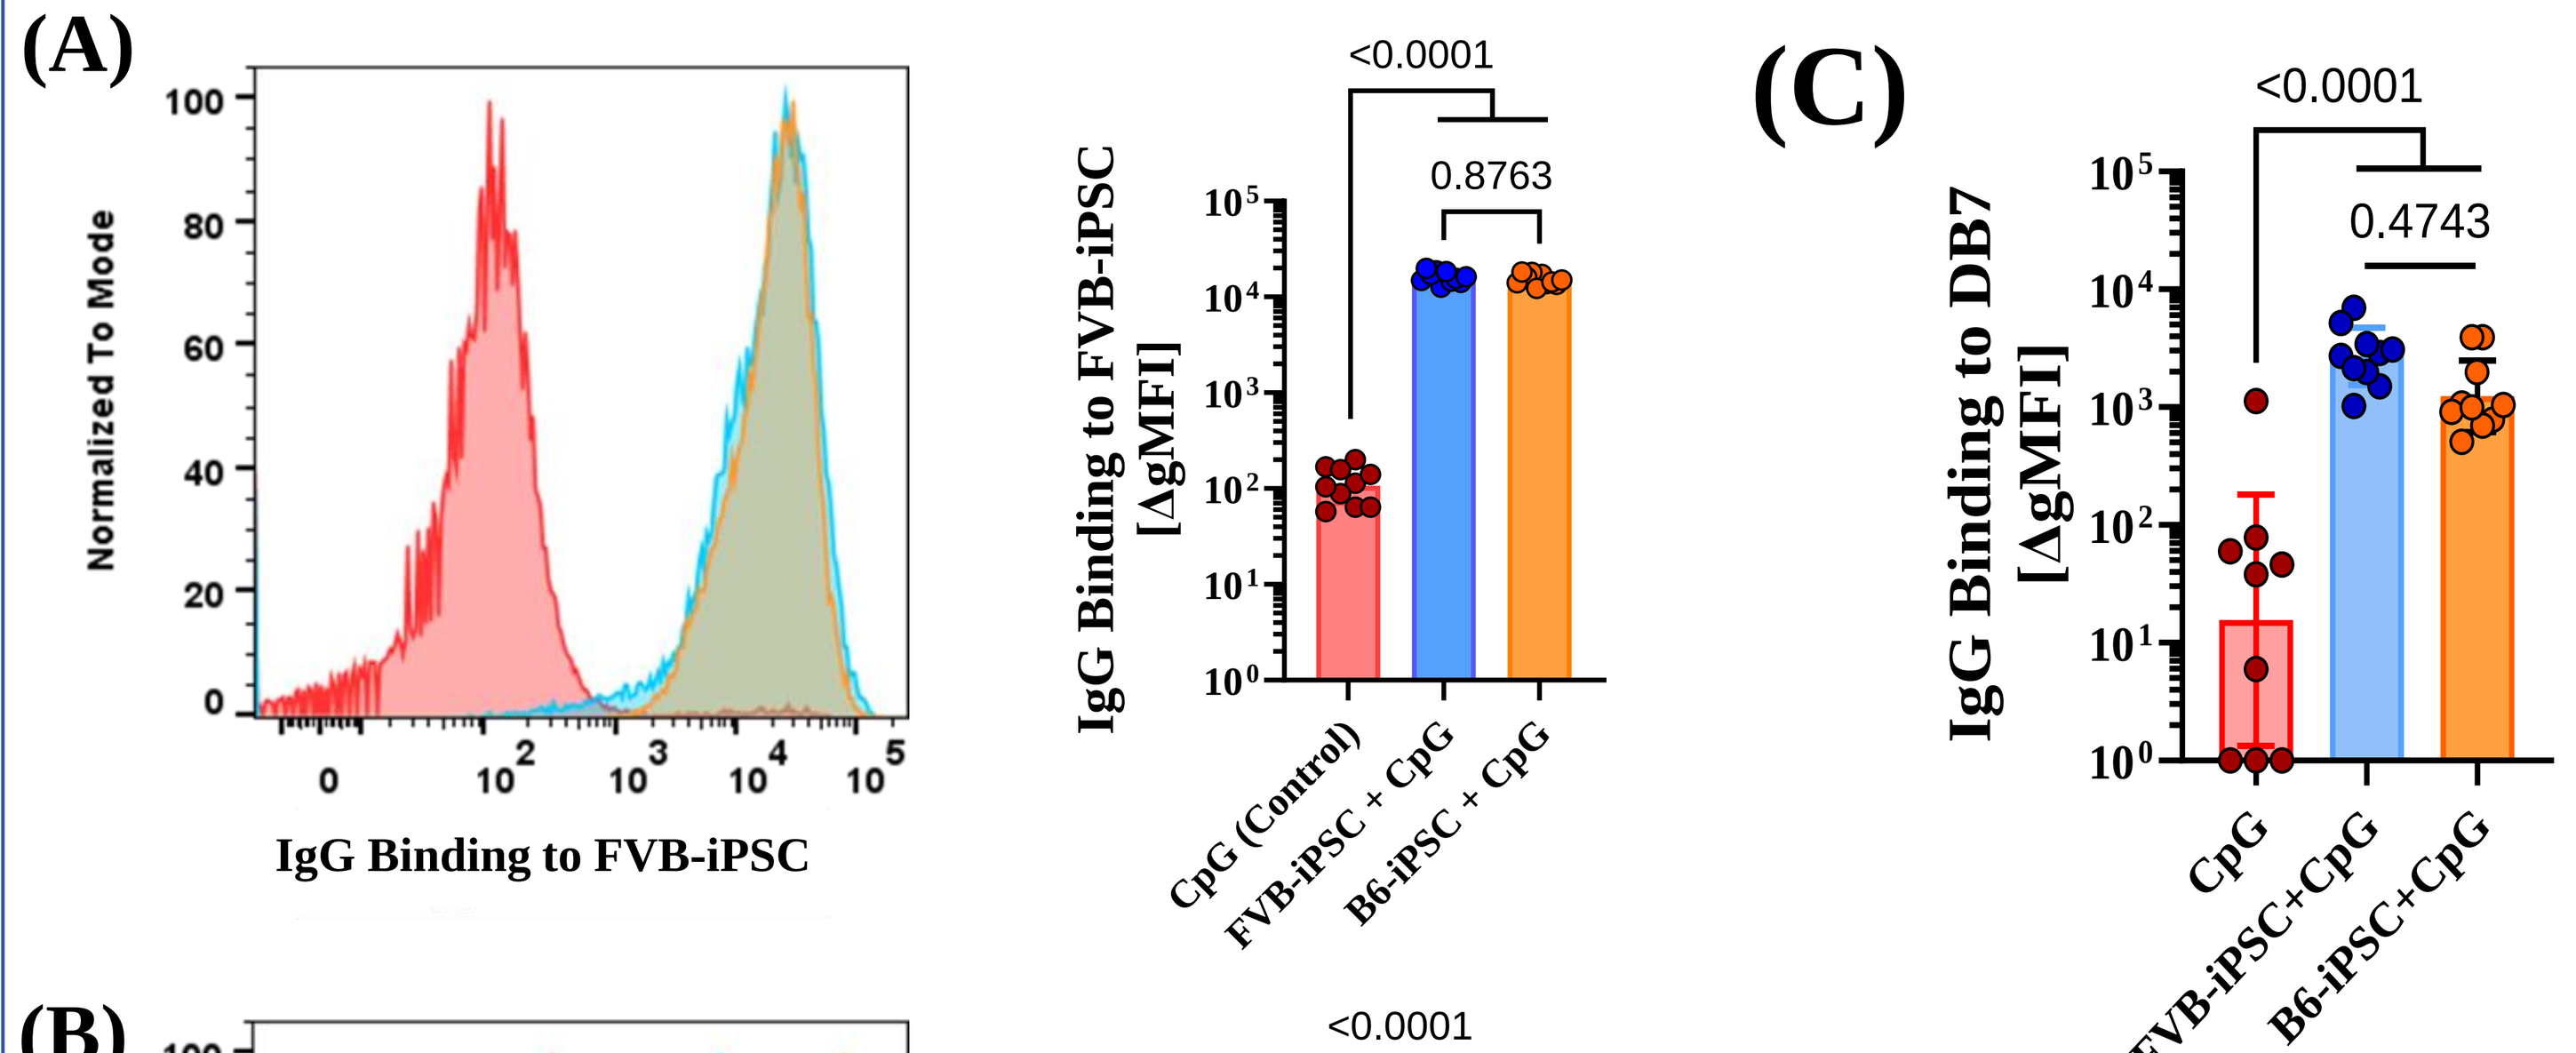
(A)
Normalized To Mode
IgG Binding to FVB-iPSC
IgG Binding to FVB-iPSC [ΔgMFI]
<0.0001
0.8763
CpG (Control)
FVB-iPSC + CpG
B6-iPSC + CpG
(C)
IgG Binding to DB7 [ΔgMFI]
<0.0001
0.4743
CpG
FVB-iPSC+CpG
B6-iPSC+CpG

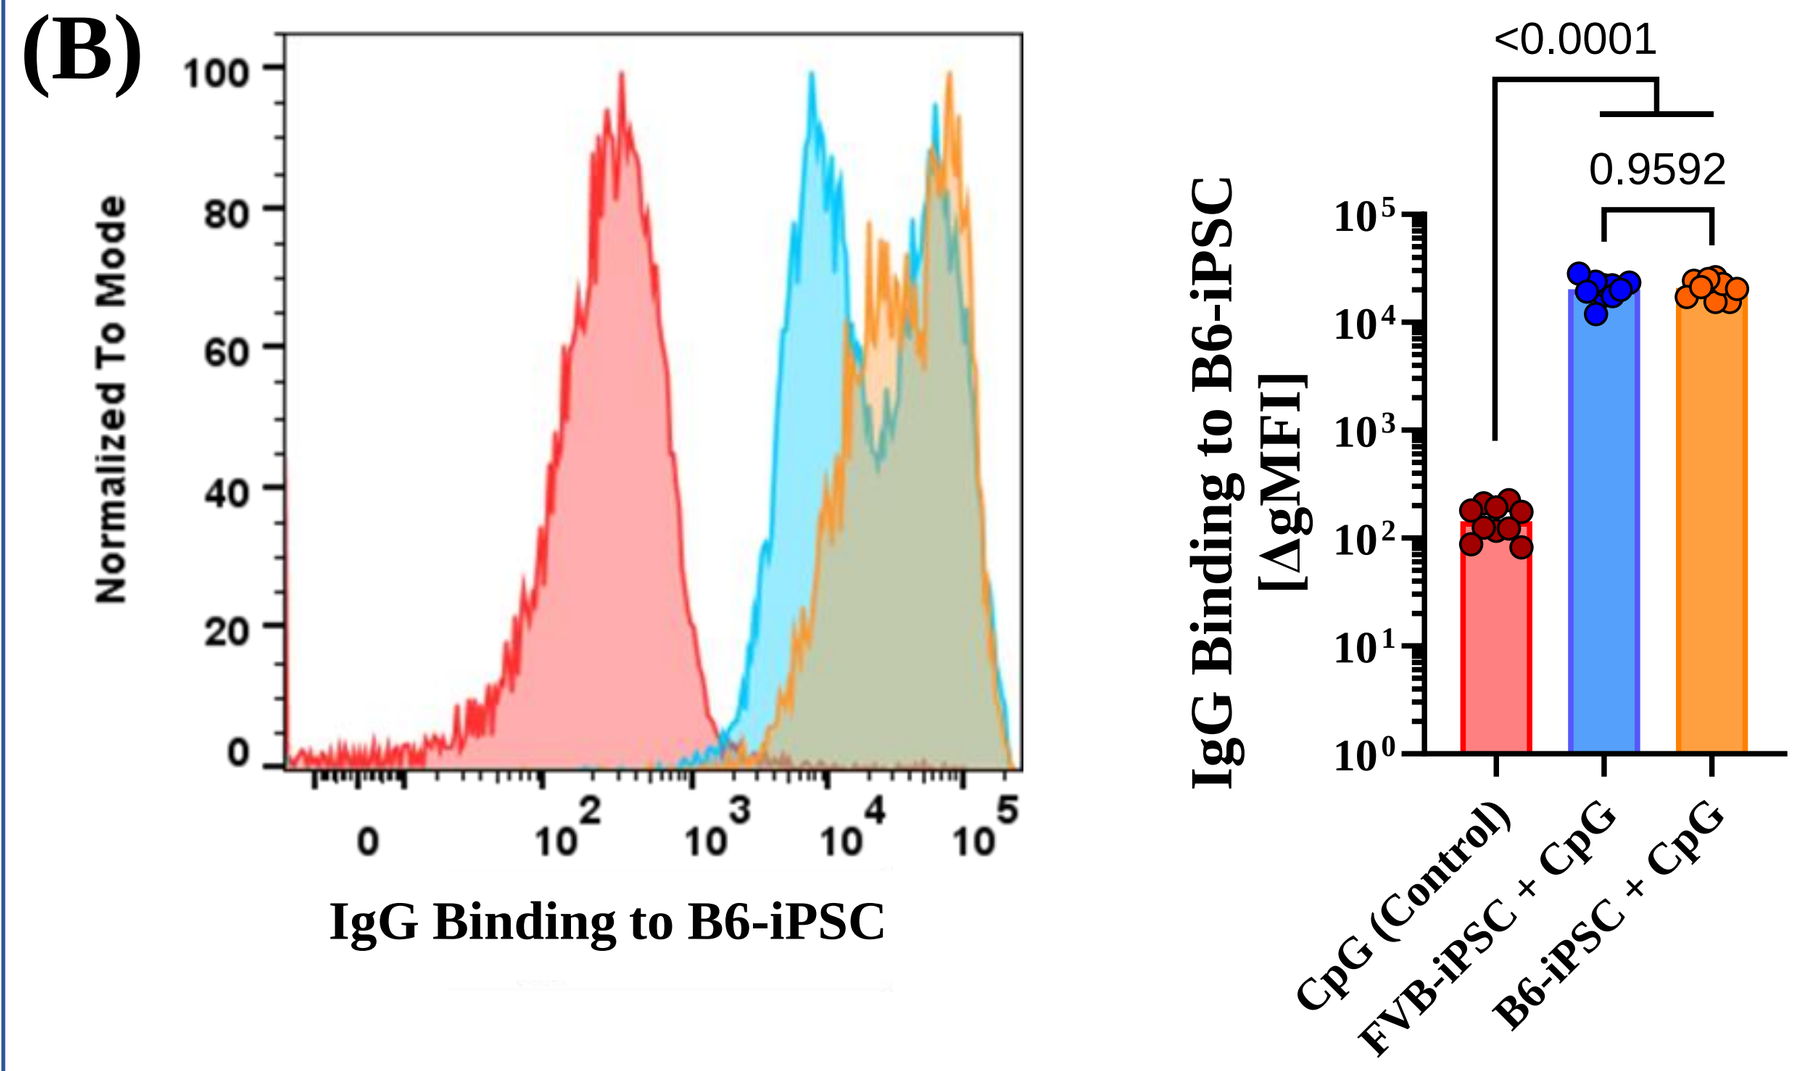
(B)
Normalized To Mode
IgG Binding to B6-iPSC
IgG Binding to B6-iPSC [ΔgMFI]
<0.0001
0.9592
CpG (Control)
FVB-iPSC + CpG
B6-iPSC + CpG

Figure 3: Mice vaccinated with syngeneic and allogeneic iPSC vaccine generate similar antibody responses. IgG binding to **(A) FVB-iPSC** and **(B) B6-iPSC** cells of sera from vaccinated mice. **(C)** IgG binding to **DB7** tumor cells of sera from vaccinated mice. (n=10, mean with SD; One-way ANOVA Tukey's test)

Cellular Response

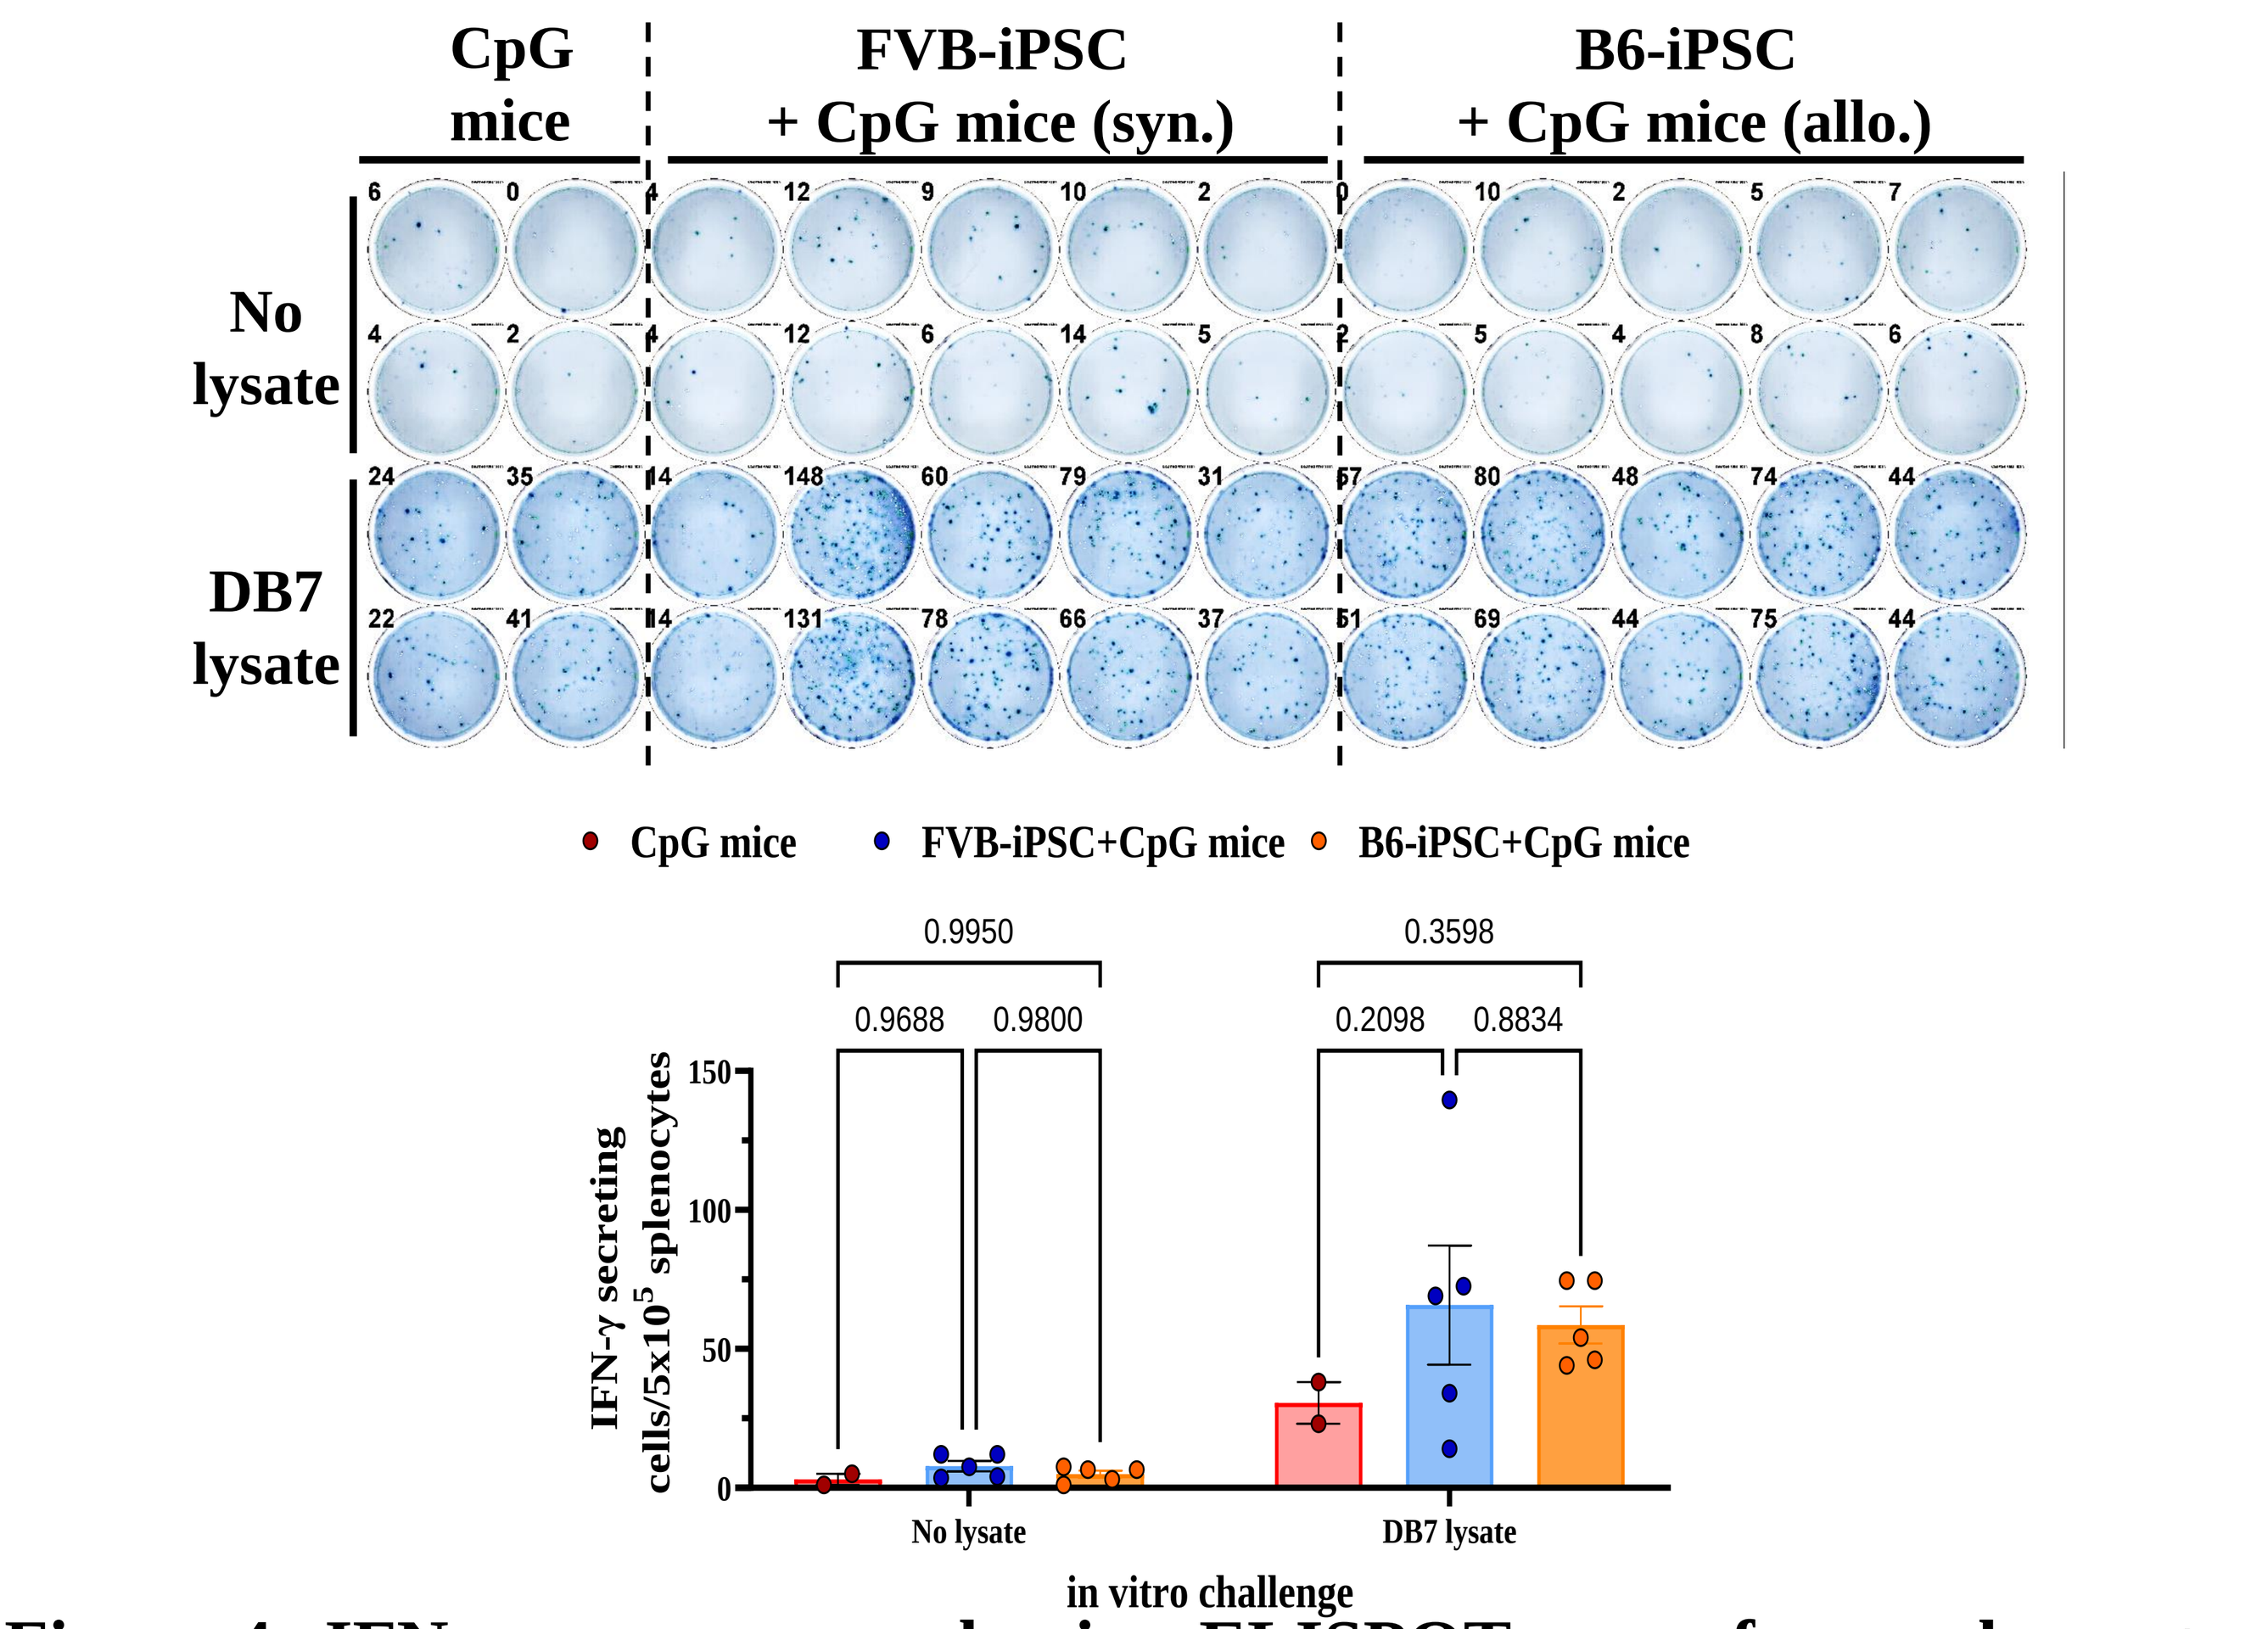
CpG mice
FVB-iPSC + CpG mice (syn.)
B6-iPSC + CpG mice (allo.)
No lysate
DB7 lysate
CpG mice
FVB-iPSC+CpG mice
B6-iPSC+CpG mice
0.9950
0.9688
0.9800
0.3598
0.2098
0.8834
IFN-γ secreting cells/5x10⁵ splenocytes
No lysate
DB7 lysate
in vitro challenge

Figure 4: IFN-γ was measured using ELISPOT assay from splenocytes isolated from vaccinated mice. (n=2 or 5, mean with SEM; Two-way ANOVA, Tukey's)

T cell (*ex vivo*)

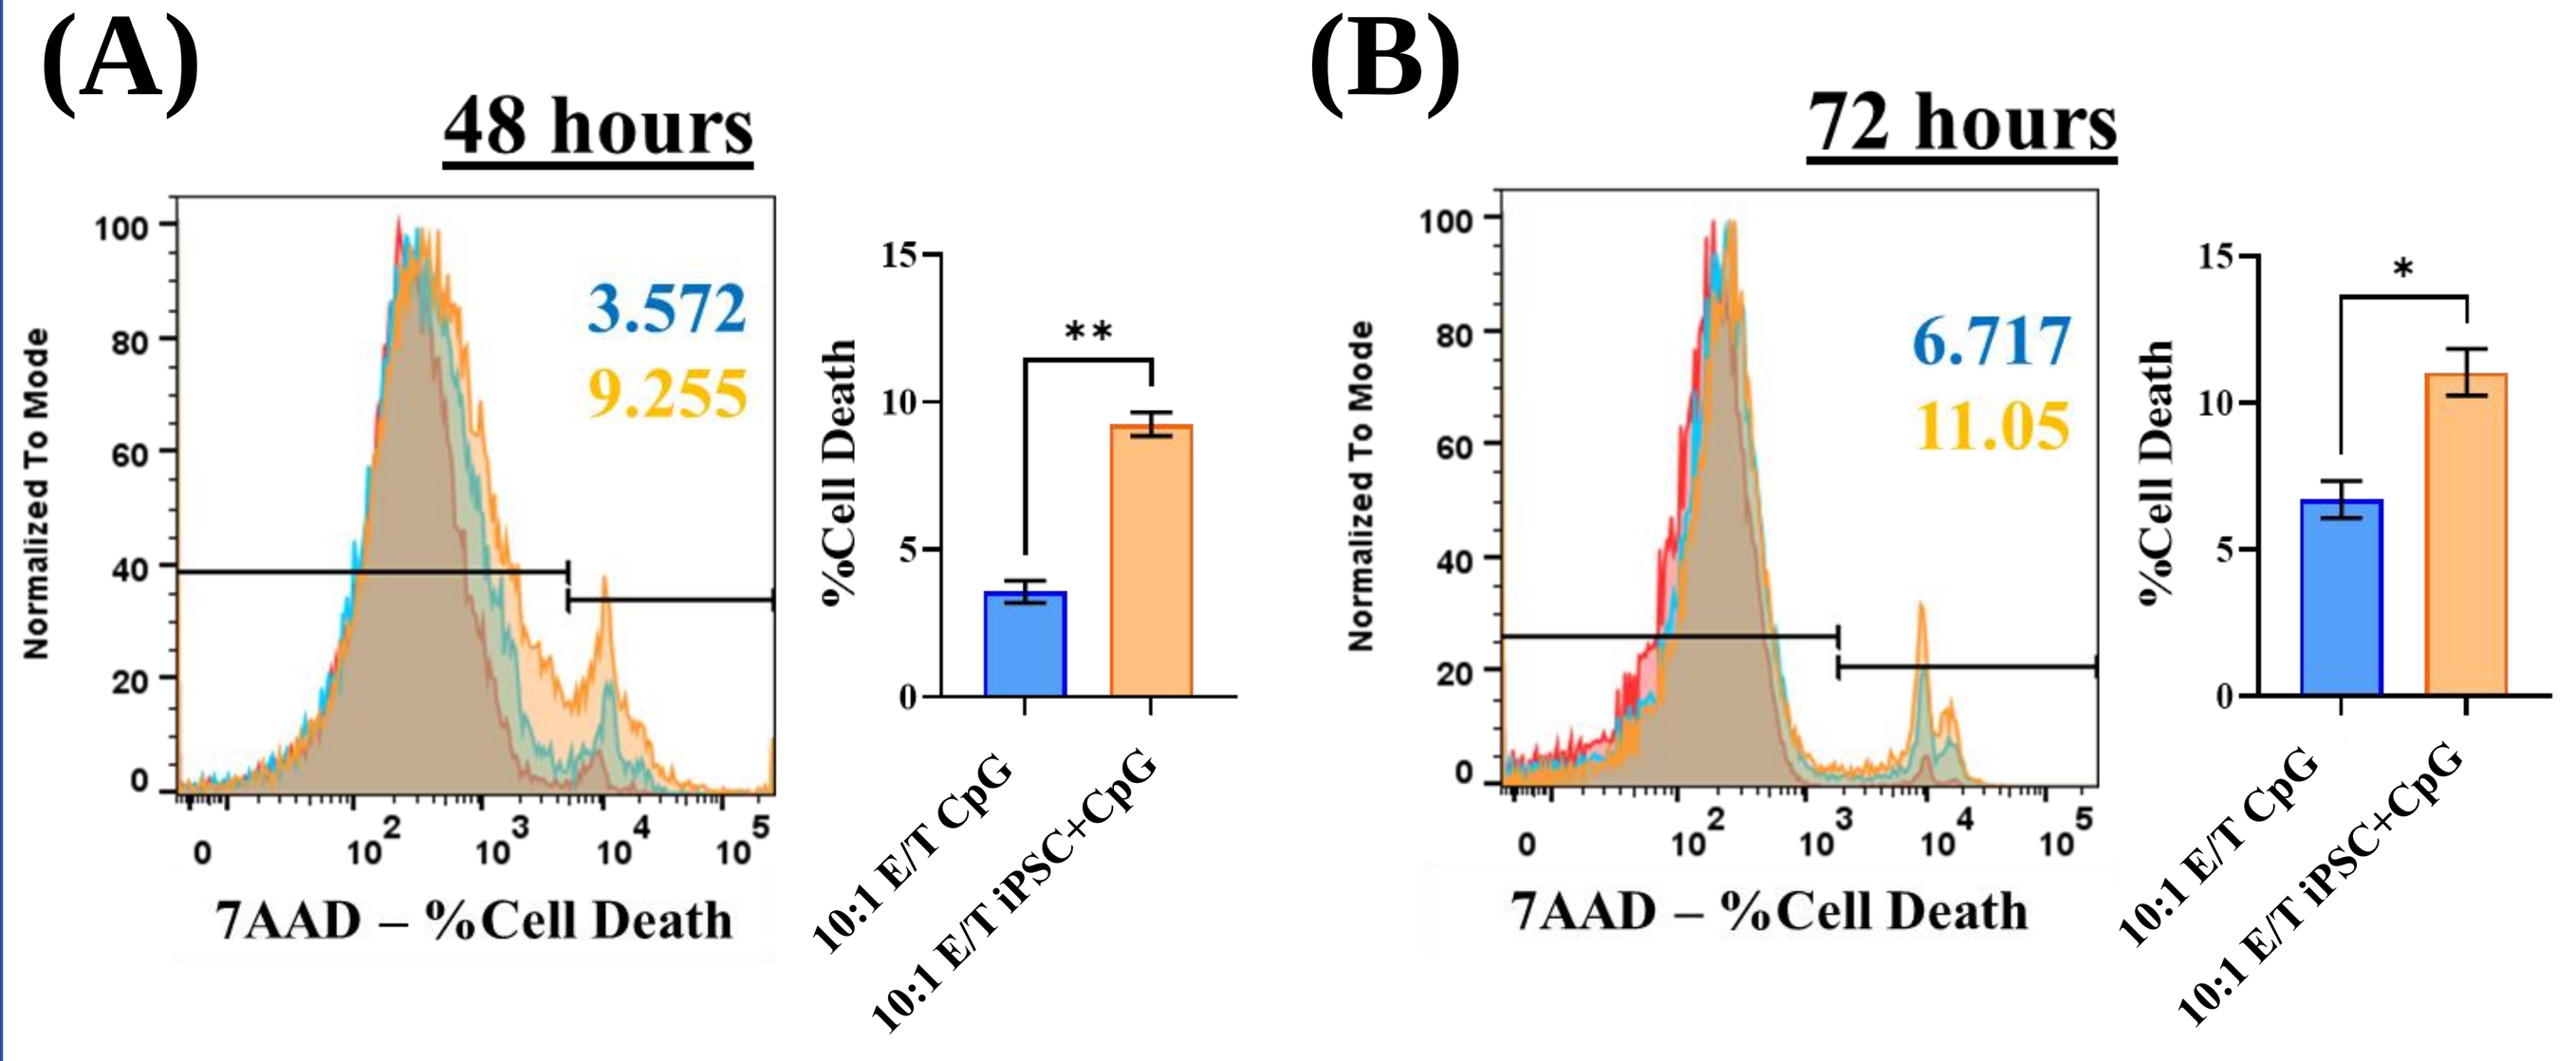
(A)
48 hours
3.572
9.255
Normalized To Mode
7AAD – %Cell Death
%Cell Death
**
10:1 E/T CpG
10:1 E/T iPSC+CpG
(B)
72 hours
6.717
11.05
*

Figure 5: Mice treated with iPSC vaccine generate significantly higher cytotoxicity response as compared to CpG control. Cytotoxicity of isolated splenic T cells against DB7 measured using flow cytometry after 48 h **(A)** and 72 h **(B)** (n=3; mean with SD; student's t-test).

Universal Vaccine

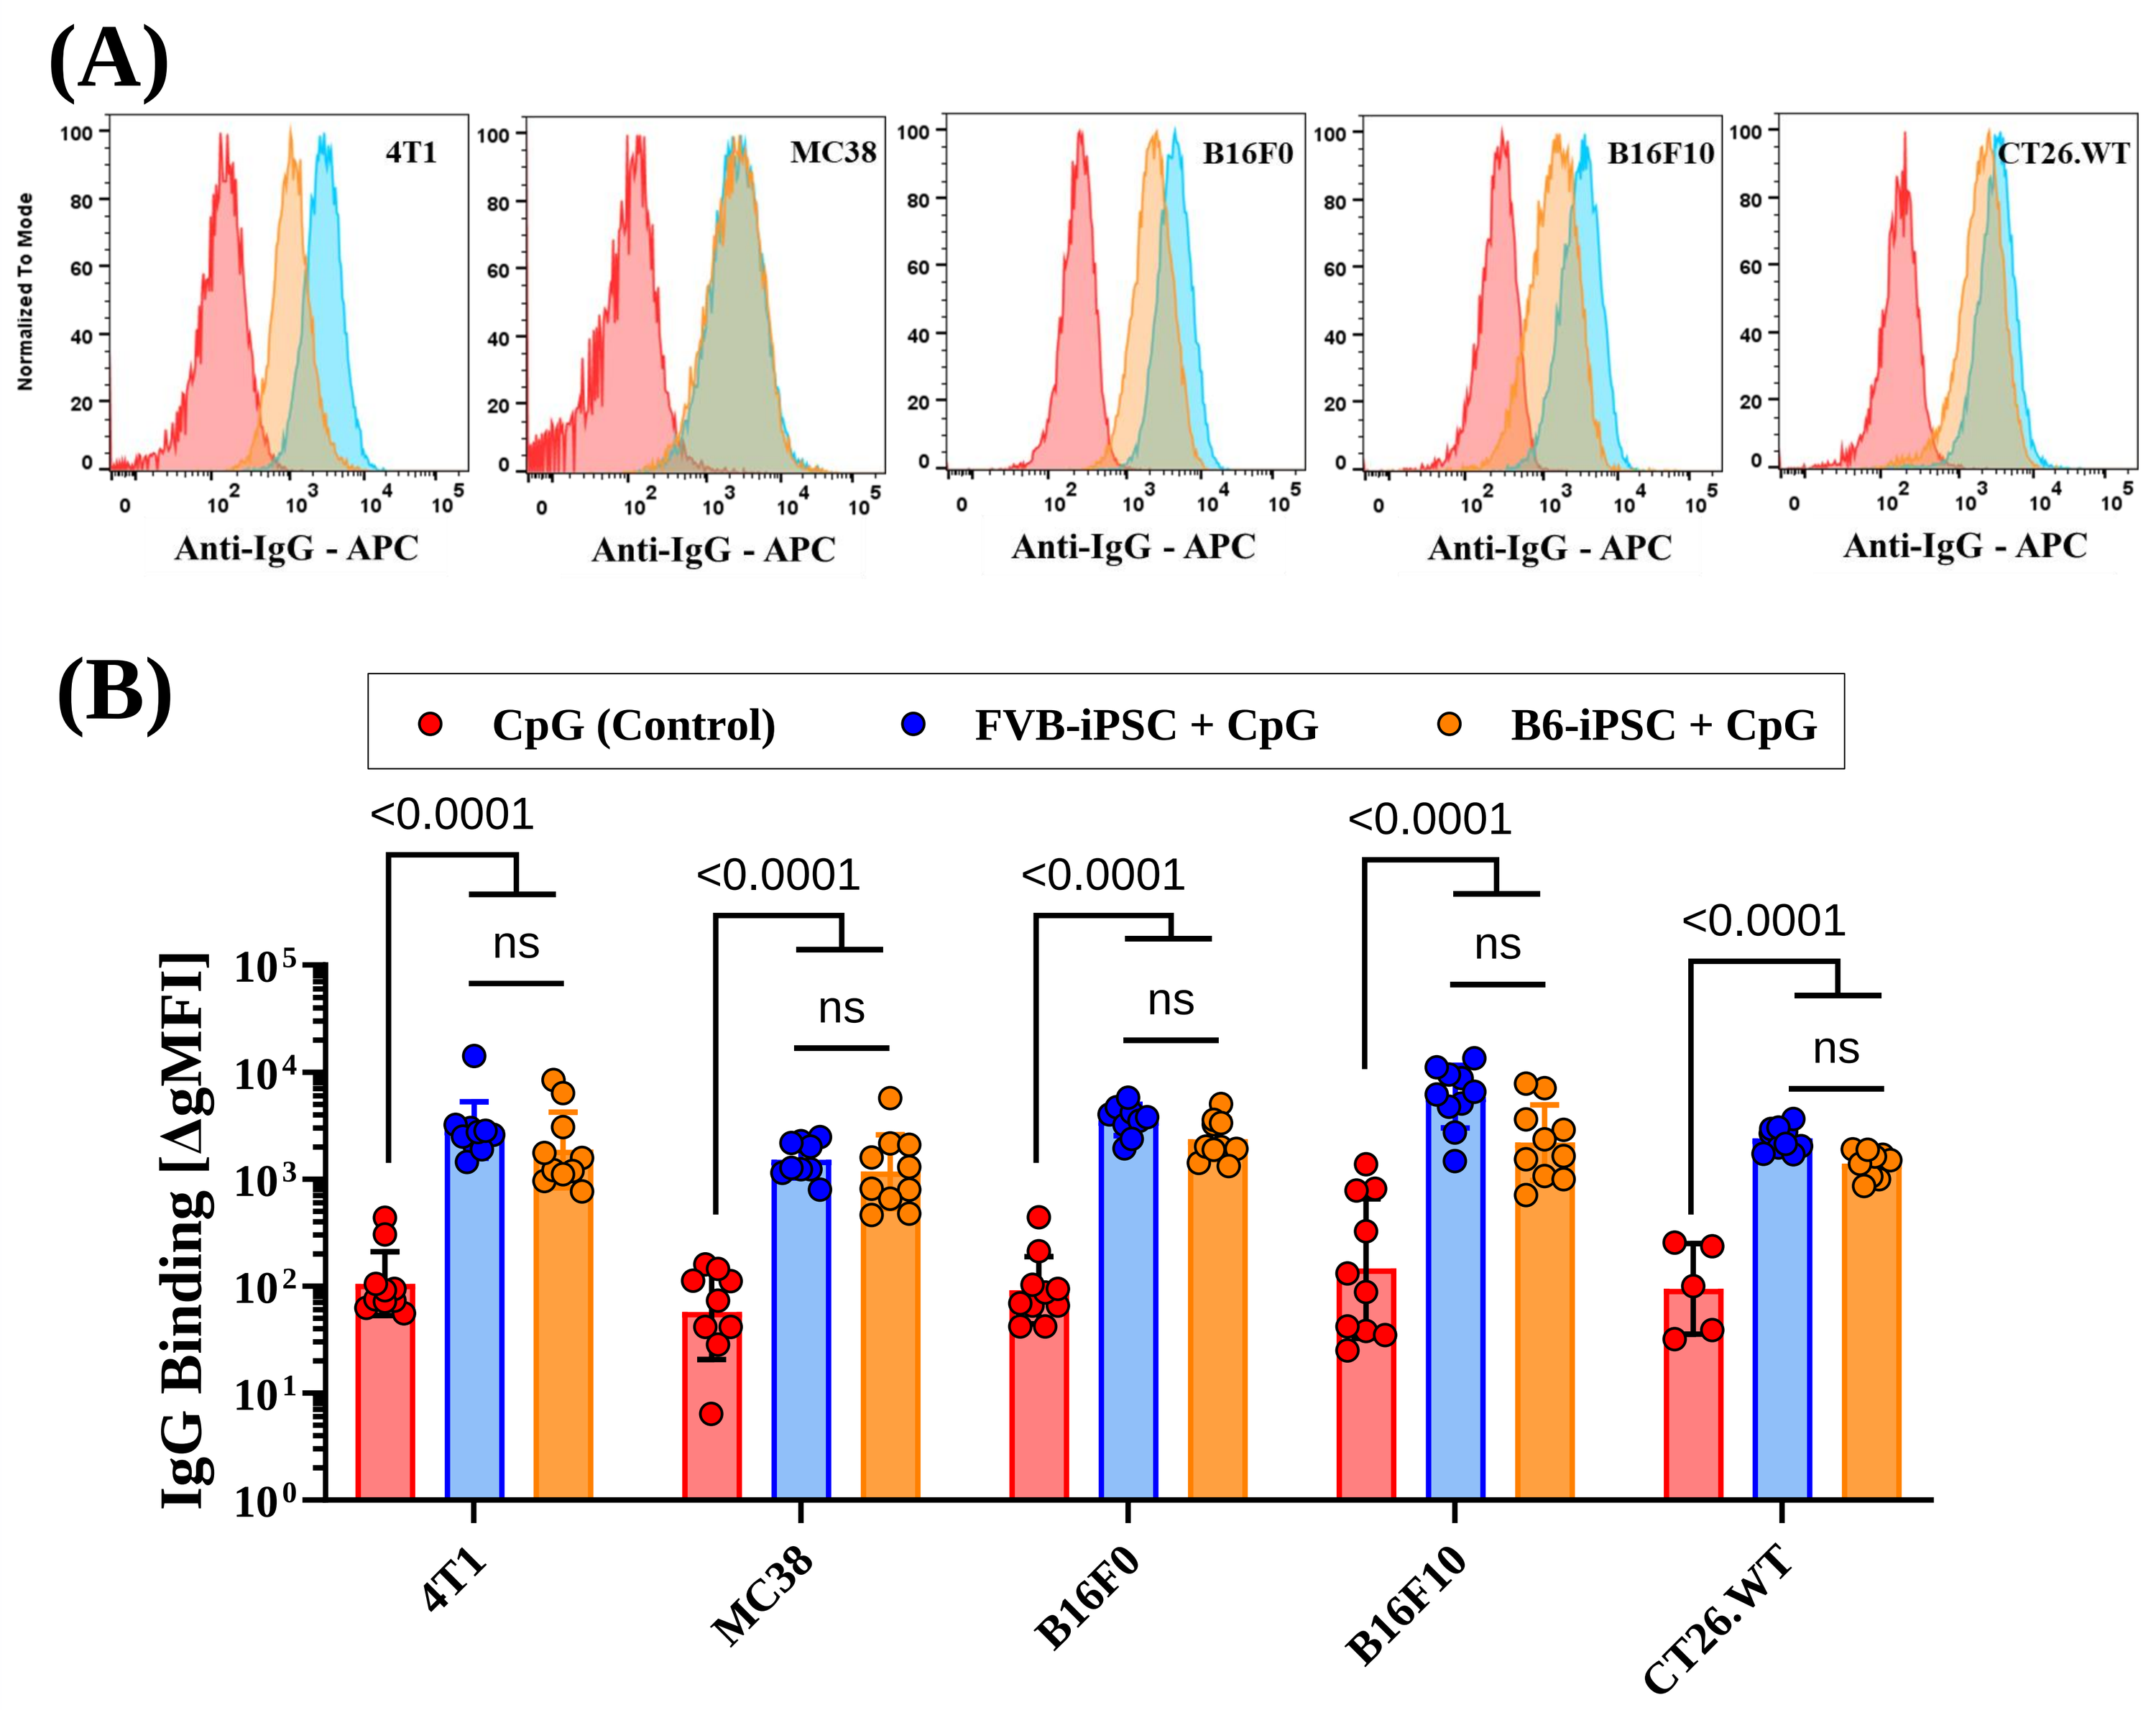
(A)
4T1
MC38
B16F0
B16F10
CT26.WT
Normalized To Mode
Anti-IgG - APC
(B)
CpG (Control)
FVB-iPSC + CpG
B6-iPSC + CpG
<0.0001
ns
IgG Binding [ΔgMFI]
4T1
MC38
B16F0
B16F10
CT26.WT

Figure 6: Allogeneic and syngeneic iPSC vaccines generate similar antibody responses against a variety of different cancer cell lines.
(A) Representative histograms of serum IgG binding from FVB- and B6-iPSC vaccinated mice to five different cancer cells lines. **(B)** Significantly higher IgG binding was observed from mice treated with FVB-iPSC and B6-iPSC vaccine as compared to CpG control mice. No differences between allogeneic and syngeneic iPSC vaccines. (n=10, mean with SD, Two-way ANOVA)

Conclusion

- Irradiated allogeneic and syngeneic iPSC admixed with TLR9 agonist CpG1826 were able to significantly delay tumor growth in DB7 injected FVB mice and induced similar antibody responses to iPSC, DB7 and other cancer cell lines.
- Allogeneic iPSC were as effective as syngeneic iPSC in murine breast cancer model.
- This result indicates the versatility of iPSC.
- Importantly, this study highlights the option of allogeneic-iPSC as a cancer treatment modality and its potential applications as a universal cancer vaccine.

Acknowledgement

The study was approved by Valley Bio Services' Institutional Animal Care and Use Committee; approval number VBS1002.

Contact

Sahil Inamdar, PhD || Scientist || sinamdar@khlorisbiosciences.com
Matthias Hundt, MD || Executive Director || mhundt@khlorisbiosciences.com

Khloris Biosciences

5729 Fontanoso Way
San Jose, CA 95138
United States

References

Kooreman NG, Kim Y, de Almeida PE, et al. Autologous iPSC-Based Vaccines Elicit Anti-tumor Responses In Vivo. Cell Stem Cell. 2018;22(4):501-513.e7. doi:10.1016/j.stem.2018.01.016

Wang L, Pegram MD, Wu JC. Induced pluripotent stem cells as a novel cancer vaccine. Expert Opin Biol Ther. 2019;19(11):1191-1197. doi:10.1080/14712598.2019.1650909

Ouyang X, Liu Y, Zhou Y, et al. Antitumor effects of iPSC-based cancer vaccine in pancreatic cancer. Stem Cell Reports. 2021;16(6):1468-1477. doi:10.1016/j.stemcr.2021.04.004

Ghosh Z, Huang M, Hu S, Wilson KD, Dey D, Wu JC. Dissecting the oncogenic and tumorigenic potential of differentiated human induced pluripotent stem cells and human embryonic stem cells. Cancer Res. 2011;71(14):5030-5039. doi:10.1158/0008-5472.CAN-10-4402